Presented at The Symposium on Advanced Wound Care, October 16–18, 2014; Las Vegas, NV

Evidence-Based Quality Improvement Initiative and Nursing/Physical Therapy Collaboration Results in Decreased Hospital-Acquired Heel Pressure Ulcers

Frances M. Dyckman, MSN, BSN, PHN, APRN-CNS, CWOCN; Christine Love, PT, DPT

ARROWHEAD REGIONAL MEDICAL CENTER

BACKGROUND

The Triple Aim is a national quality strategy that has gained much attention since the Affordable Care Act (ACA) legislation leveraged this concept in 2010. The 3 primary aims of the Triple Aim are:[1]

"Better Care: Improve the overall quality, by making health care more patient-centered, accessible, and safe."

"Healthy People/Healthy Communities: Improve the health of the U.S. population by supporting proven interventions to address behavioral, social and, environmental determinants of health in addition to delivering higher-quality care."

"Affordable Care: Reduce the cost of quality health care for individuals, families, employers, and government."

The prevention of hospital-acquired heel pressure ulcers (HAhPUs) is a major focus of the Triple Aim. The development of HAhPUs is associated with increased patient morbidity and pain, decreased quality of life, extended hospital length of stay, and increased costs.[2] The heel is the second most prevalent anatomic location for pressure-related breakdown.[3]

Evidence-based guidance has been published on HAhPU prevention, which consists of appropriate heel offloading.[4] Although there is no consensus on the most effective heel-offloading device, a device should ensure that leg weight is redistributed along the calf without undue pressure on the Achilles tendon, effectively floating the heel off the surface while immobile, and preventing foot drop.[5]

Public hospitals are faced with patients with complex issues, some of whom are homeless, living in poverty, and/or have high risk factors for HAhPU development. This quality assurance/performance improvement (QAPI) intervention was implemented to meet the objectives of the Triple Aim, reduce the incidence of HAhPUs, and improve patient outcomes.

METHODS

ANALYSES: Past, present, and future methodologic analyses and comparisons were conducted to help identify strengths and weaknesses that needed to be addressed for HAhPU prevention.

Risk assessment protocols and prevention guidelines were not broadly understood and accepted by staff. In addition the prevention of the adverse events of HAhPU and plantar flexion contractures (foot drop) were not a priority for the staff. There was a need for evidence-based education on how to appropriately identify patients at risk for HAhPU development and how to implement risk-stratified interventions based upon Braden Risk Assessment Scoring.

It was determined that after the return to the original heel offloading device, an algorithm standardizing application criteria for device would be used for interprofessional education and to facilitate collaboration in all efforts to prevent HAhPU.

EVALUATION: The baseline HAhPU rate was calculated and compared with the past intervention rate. Each heel ulcer was analized using a root-cause analysis process to determine the gaps in care. After determination of the gaps, a PDSA (Plan Do Study Act) performance improvement analysis supported the need for the change in process and intervention. It should be noted that the heel offloading device utilized for HAhPU prevention had been changed to a less expensive option between November 2011 to February 2012. The intervention focused on product selection with functional criteria to include heel off-loading for extreme bariatric patients and prevention of plantar flexion contractures, and the heel off-loading device used prior to November 2011 was re-implemented. Post PDSA and intervention initiation, a caregiver perception survey was administered to the Physical Therapy and Nursing staff. This survey was designed to assess the competency with risk assessment and the perception of the priority for prevention of HAhPU and plantar flexion contracture prevention. Interventions:

- Heel off-loading device* changed
- Nursing education to risk assessment and proper device application
- Physical therapy education to use of algorithm for heel pressure relief
- Coordination of interprofessional communications with regular meetings
- Algorithm development for evidence-based HAhPU prevention
- Incorporation of heel pressure relief algorithm to standardize of evidence-based bundles of care

*Prevalon® Pressure-Relieving Heel Protector (Sage Products LLC, Cary, IL)

METHODS continued

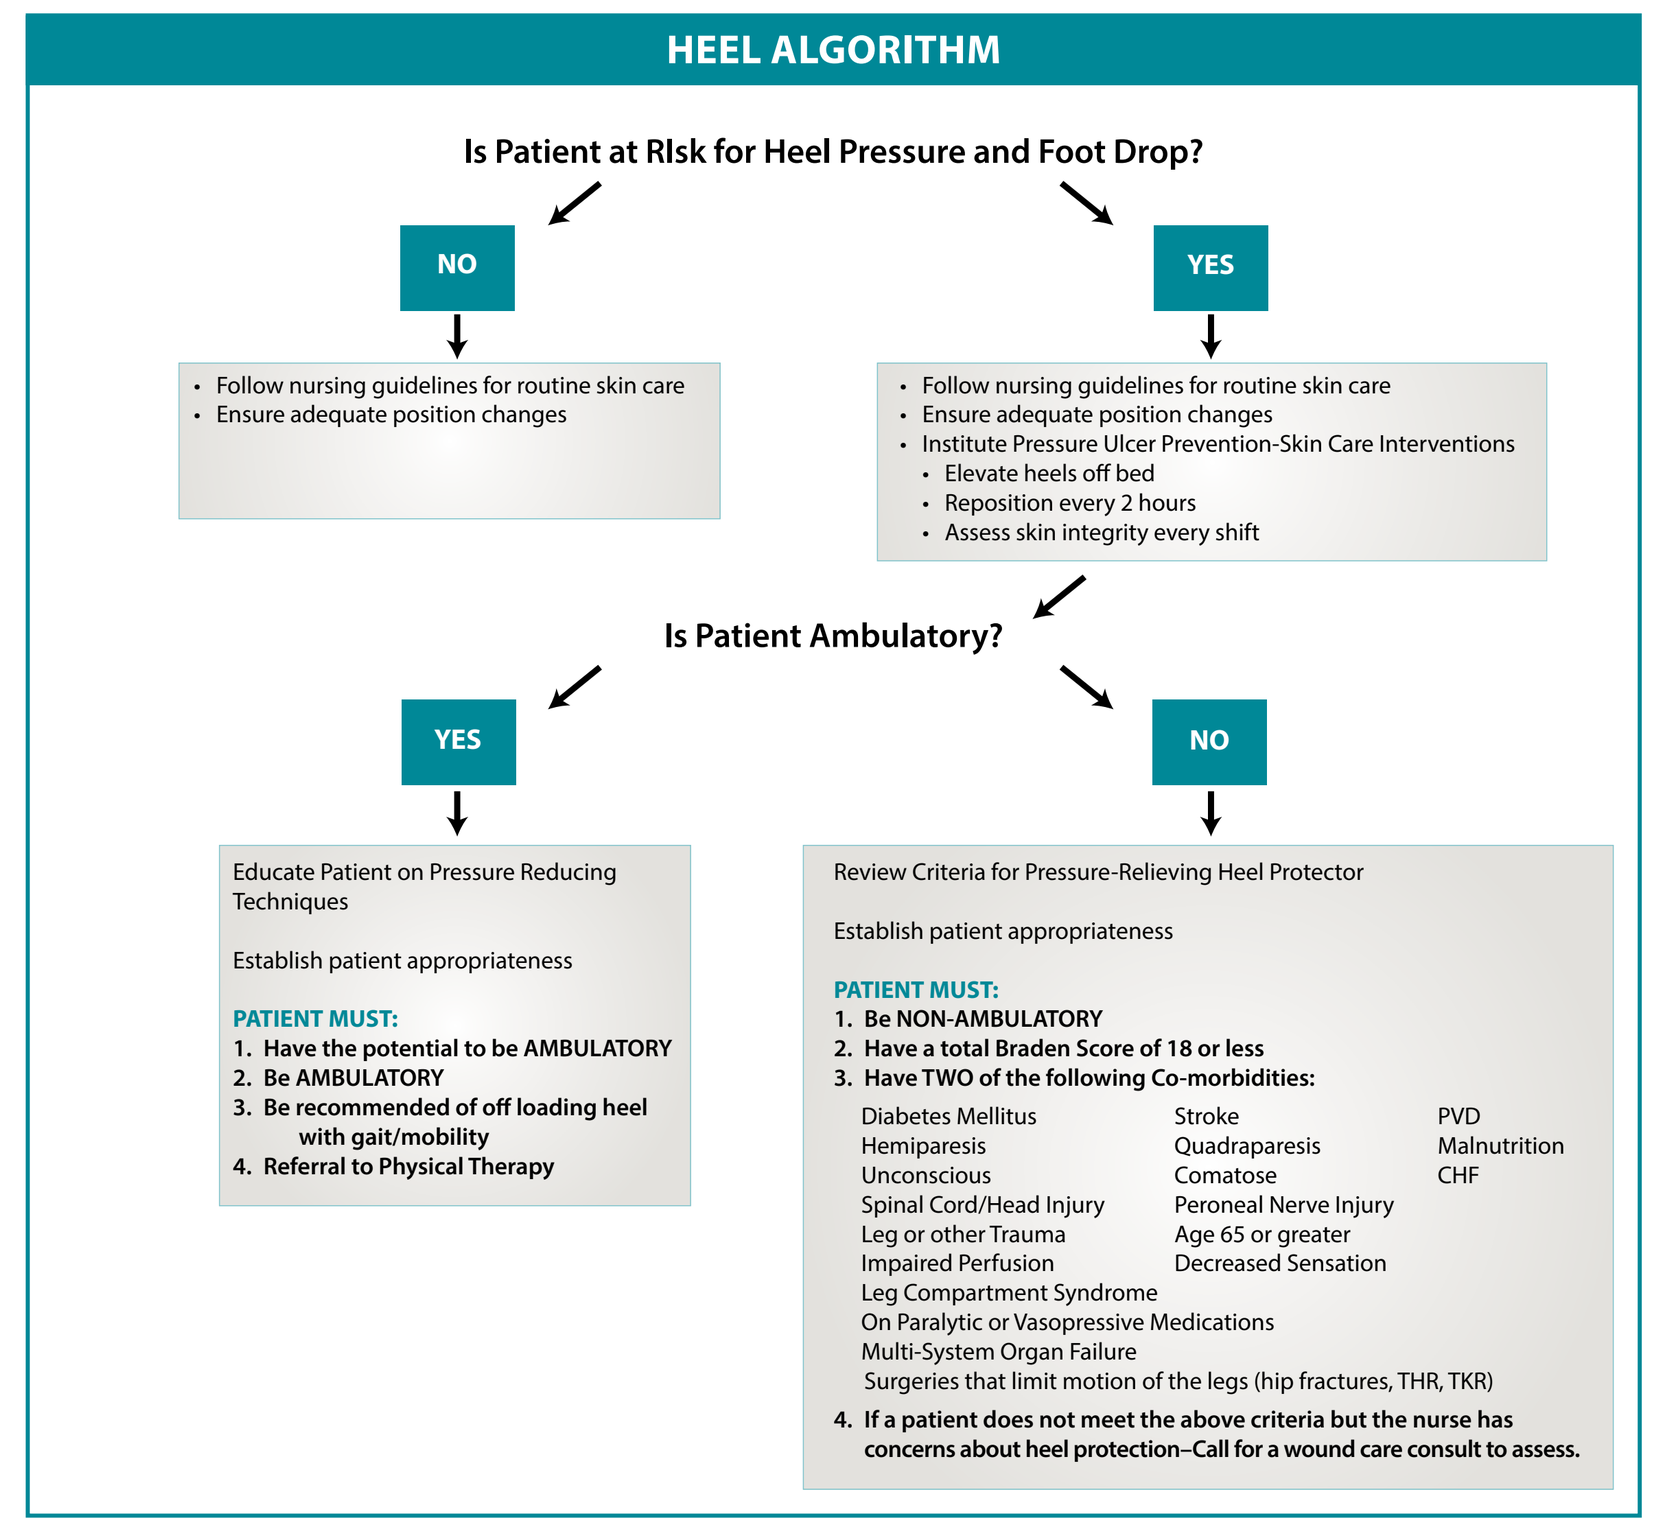

HEEL ALGORITHM
Is Patient at RIsk for Heel Pressure and Foot Drop?
NO
YES
• Follow nursing guidelines for routine skin care
• Ensure adequate position changes
• Follow nursing guidelines for routine skin care
• Ensure adequate position changes
• Institute Pressure Ulcer Prevention-Skin Care Interventions
• Elevate heels off bed
• Reposition every 2 hours
• Assess skin integrity every shift
Is Patient Ambulatory?
YES
NO
Educate Patient on Pressure Reducing Techniques
Establish patient appropriateness
PATIENT MUST:
1. Have the potential to be AMBULATORY
2. Be AMBULATORY
3. Be recommended of off loading heel with gait/mobility
4. Referral to Physical Therapy
Review Criteria for Pressure-Relieving Heel Protector
Establish patient appropriateness
PATIENT MUST:
1. Be NON-AMBULATORY
2. Have a total Braden Score of 18 or less
3. Have TWO of the following Co-morbidities:
Diabetes Mellitus
Hemiparesis
Unconscious
Spinal Cord/Head Injury
Leg or other Trauma
Impaired Perfusion
Leg Compartment Syndrome
On Paralytic or Vasopressive Medications
Multi-System Organ Failure
Surgeries that limit motion of the legs (hip fractures, THR, TKR)
Stroke
Quadraparesis
Comatose
Peroneal Nerve Injury
Age 65 or greater
Decreased Sensation
PVD
Malnutrition
CHF
4. If a patient does not meet the above criteria but the nurse has concerns about heel protection–Call for a wound care consult to assess.

RESULTS

The QAPI intervention was deemed successful after an initial before-after review of HAhPU rates, which showed an approximate 70.6% reduction of HAhPUs. A sustained improvement required focused education and competency checks during the March 2012 house-wide nursing skills day. Inclusion of this competency check on a regular basis has ensured a consistent reduction in our HAhPU rate in the post-intervention period.

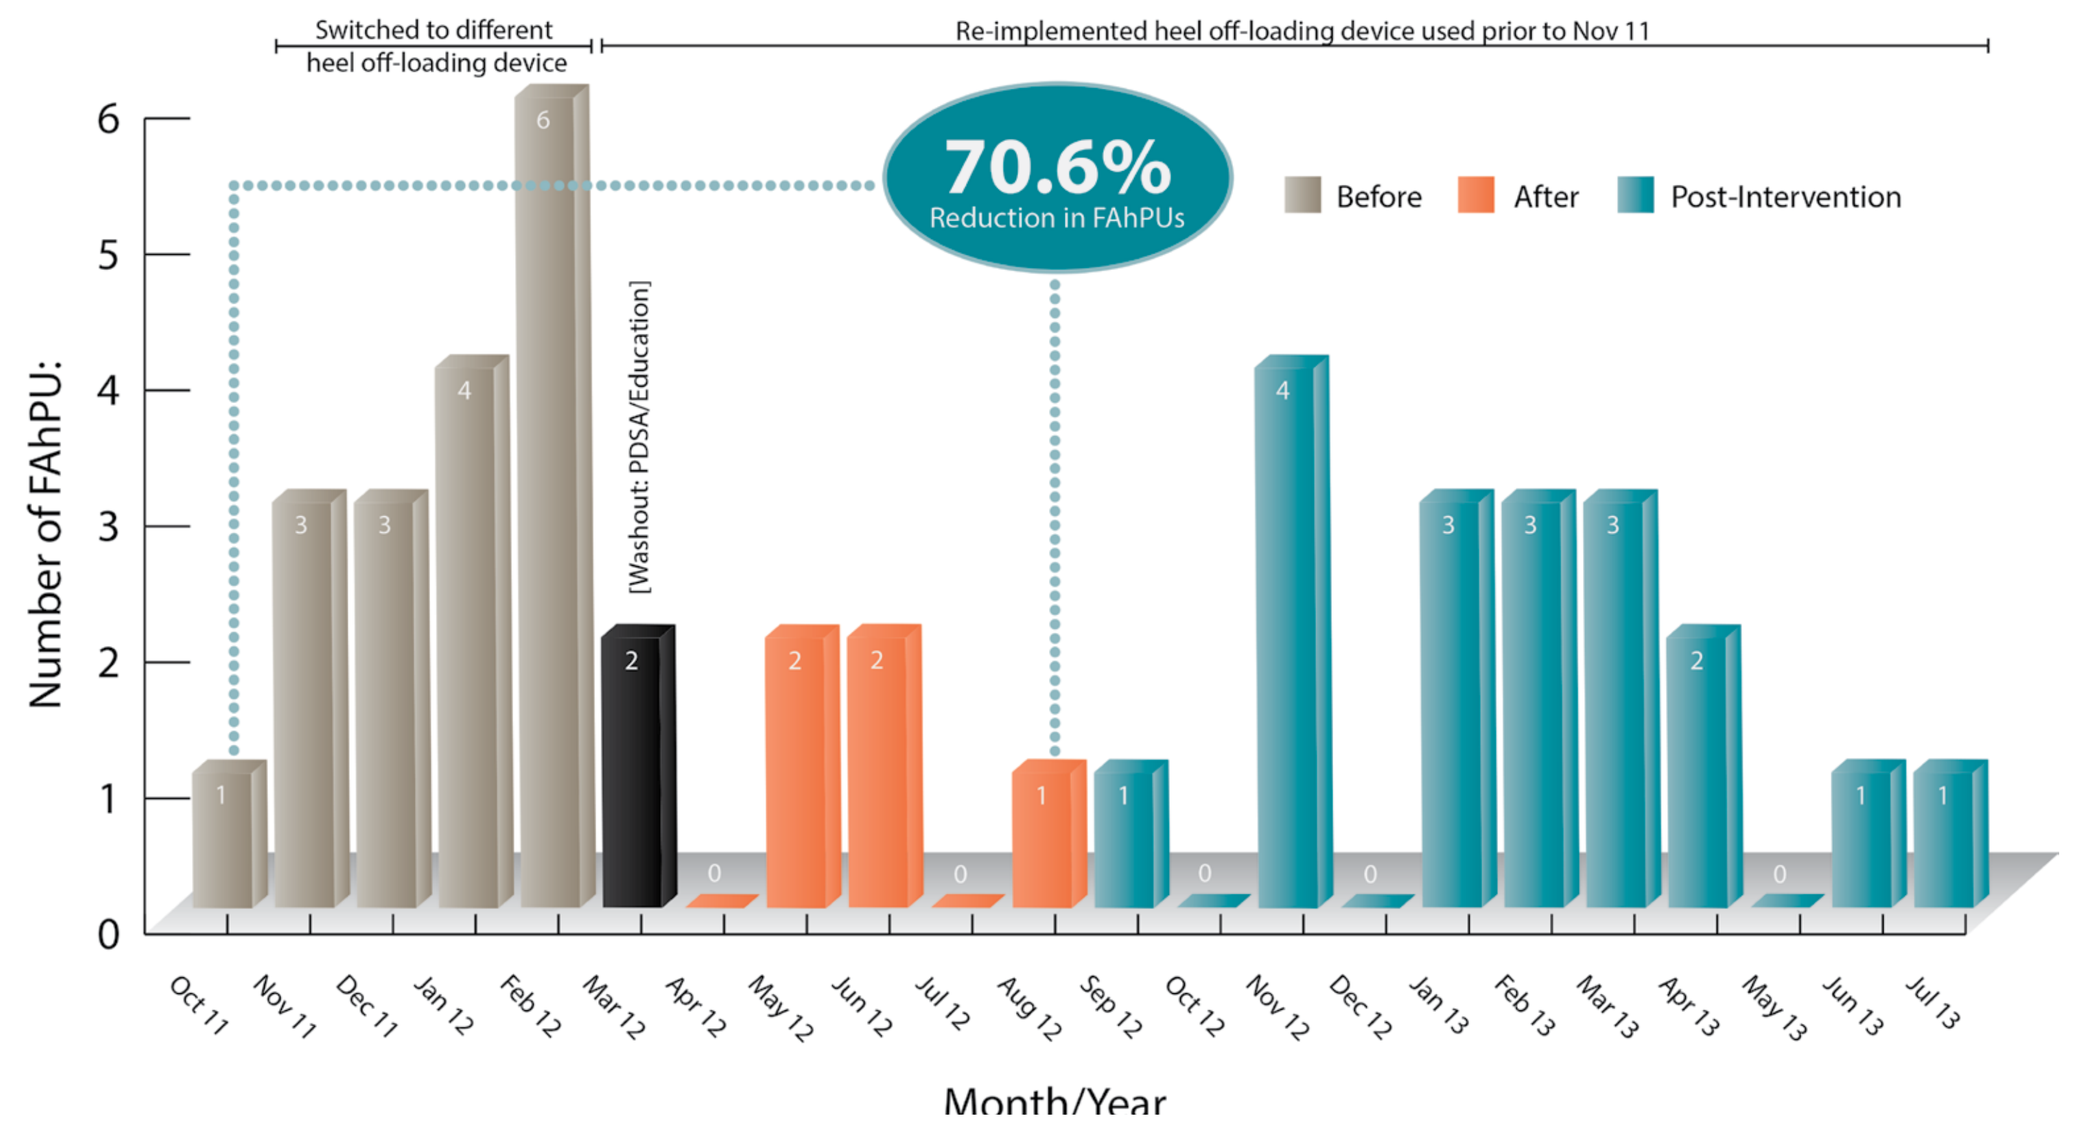

Switched to different heel off-loading device
Re-implemented heel off-loading device used prior to Nov 11
70.6%
Reduction in FAhPUs
Before
After
Post-Intervention
[Washout: PDSA/Education]
Number of FAhPU:
Month/Year
Oct 11
Nov 11
Dec 11
Jan 12
Feb 12
Mar 12
Apr 12
May 12
Jun 12
Jul 12
Aug 12
Sep 12
Oct 12
Nov 12
Dec 12
Jan 13
Feb 13
Mar 13
Apr 13
May 13
Jun 13
Jul 13

CLINICAL IMPLICATIONS

The implications of this successful project are wide ranging for public hospitals and other hospital systems. By driving best practices to the patient's bedside and enhancing collaboration between nursing and physical therapy staff, we have improved patient outcomes, staff education and competencies, patient quality of care, and decreased excess costs.

A cross-discipline, respectful relationship was forged during our QAPI experience. It was recognized that the input of an expert clinician is necessary during the decision-making process for product changes. Prevention products are now recognized as important components for patient safety and the avoidance of adverse events.

Although costs are difficult to justify based on cost avoidance, the fiscal and materials management department now recognizes the contribution of continued analysis of best practices and dollars spent in prevention versus dollars lost after a negative avoidable event such as HAhPU development.

REFERENCES

1. Berwick DM, Nolan TW, Wittington J. The Triple Aim: Care, Health, and Cost. Available at: http://content.healthaffairs.org/content/27/3/759. abstract
2. Sullivan N, Schoelles KM. Preventing in-facility pressure ulcers as a patient afety strategy: a systematic review. Ann Intern Med. 2013;158:410-6.
3. VanGilder C, Amlung S, Harrison P, Meyer S. Results of the 2008-2009 International Pressure Ulcer Prevalence Survey and a 3-year, acute care, unit-specific analysis. Ostomy Wound Manage. 2009;55(11):39-45.
4. European Pressure Ulcer Advisory Panel and National Pressure Ulcer Advisory Panel. Prevention and treatment of pressure ulcers: quick reference guide. Washington DC: National Pressure Ulcer Advisory Panel; 2009.
5. Junkin J, Gray M. Are pressure redistribution surfaces or heel protection devices effective for preventing heel pressure ulcers? J Wound Ostomy Continence Nurs. 2009;36(6):602-8.