Presented at the 23rd Annual Meeting of the Wound Healing Society; SAWC Spring/WHS Joint Meeting: Denver, Colorado • May 1 - 5, 2013

Incontinence Associated Dermatitis in the Acute Care Setting:
A Prospective Multi-site Epidemiologic Study

Mikel Gray, PhD, FNP, PNP, CUNP, CCCN, FAANP, FAAN | University of Virginia Department of Urology | Charlottesville, VA
Steve Bartos, MS, ED | PhD Candidate | Illinois Institute of Technology | Department of Mathematics and Science Education | Chicago, IL

ABSTRACT

Introduction: Despite a growing number of studies, knowledge of the epidemiology, etiology, pathophysiology, and clinical characteristics of Incontinence Associated Dermatitis (IAD) remains limited.

Purpose: This study describes findings from a multi-site national IAD database in acute care facilities. Specific study aims were: 1) to measure the prevalence of IAD in the acute care setting, 2) to describe clinical characteristics of IAD, and 3) to analyze the relationship between IAD and prevalence of sacral/coccygeal pressure ulcers.

Methods: IAD prevalence was measured during a single 24-hour period. Skin status, including presence of IAD and sacral/coccygeal pressure ulcers, was evaluated by skin champion nurses at each facility using a standardized form.

Results: Skin assessments were completed in 791 patients in 20 facilities throughout the United States. One hundred twenty nine patients (16.3%) were found to have perineal skin damage; 184 (23.3%) were deemed to have IAD. All patients with IAD had urinary, fecal or double incontinence. The type of incontinence influenced the likelihood of IAD. The prevalence of IAD among patients with urinary incontinence alone was 4.3%; it was 29.3% among patients with fecal incontinence and 66.3% among patients with double incontinence ($p=0.01$). IAD severity was not associated with incontinence type ($p=.329$). Slightly over a quarter (27.9%) of patients with IAD had erosion of the perineal skin, and 17% had cutaneous candidiasis. Slightly over a quarter of IAD (26%) was present on admission, and 74% was hospital acquired. IAD was associated with an increased prevalence of sacral/coccygeal pressure ulcers ($p<0.000$). No association was seen between PU stage and presence of IAD.

Conclusion: IAD is a prevalent condition among patients with urinary, fecal or double incontinence. IAD is associated with an increased likelihood of sacral/coccygeal pressure ulcers.

METHODS

Incontinence associated dermatitis (IAD) prevalence and sacral PU prevalence data were extracted from a database of point prevalence data (N=791). Descriptive statistics were provided using frequency and percentage of cohort. Missing data values are noted in tables. A digital point prevalence tool was utilized for data collection (Figure 1).

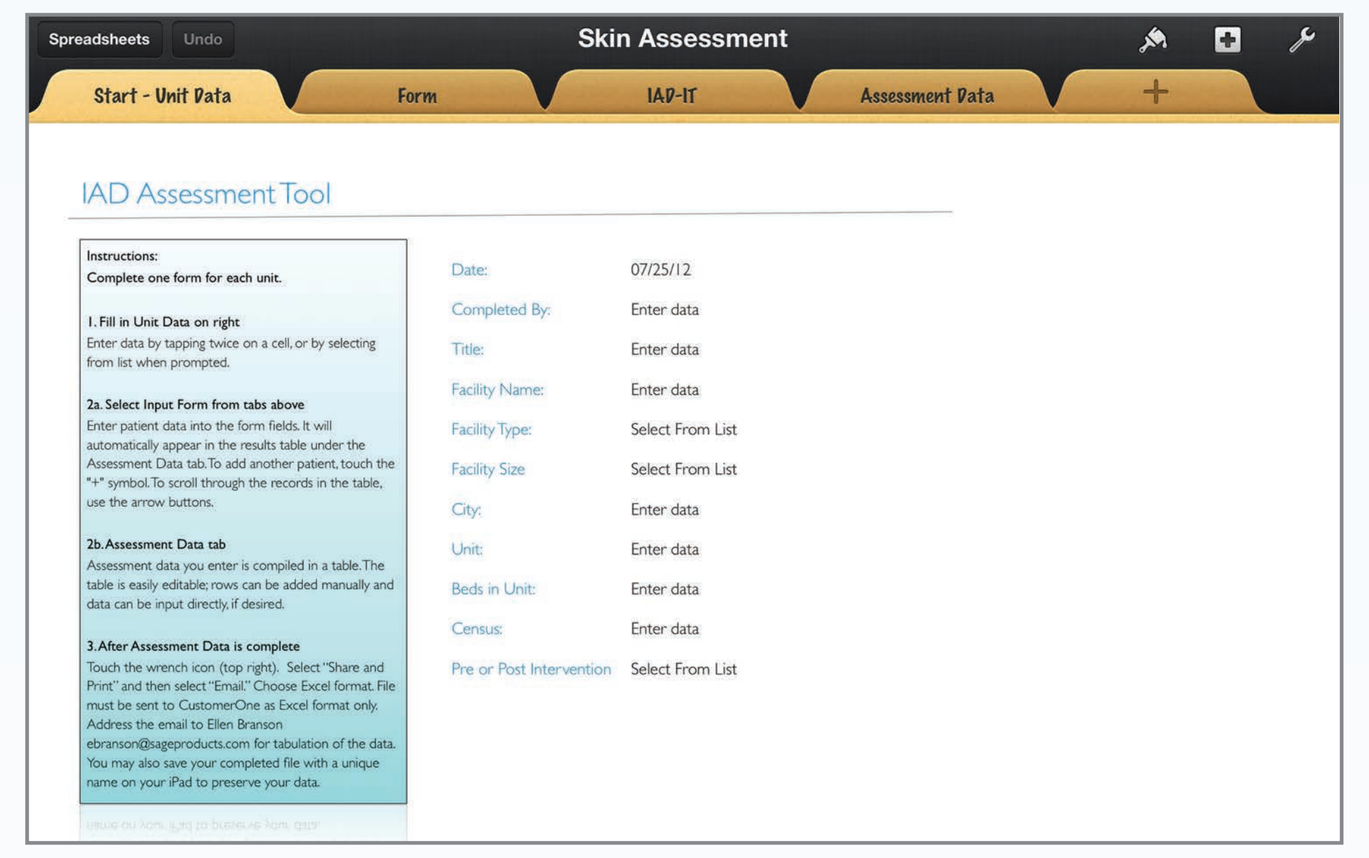
Skin Assessment
Start - Unit Data
Form
IAD-IT
Assessment Data
IAD Assessment Tool

RESULTS

Descriptive Cohort Data (N=791)
The following table (Table 1) presents descriptive statistics on the cohort data, such as facility size, location, and unit type.

Table 1. Sample characteristics and research setting

TYPE OF UNIT	Frequency	Percent
ICU	354	44.8
Other	404	51.1
Missing data	33	4.2
Total	791	100.0
FACULTY SIZE	Frequency	Percent
0 - 199 Beds	175	22.1
200 or more	564	71.3
Missing data	52	6.6
Total	791	100.0
FACULTY TYPE	Frequency	Percent
Rehab	15	1.9
Hospital	743	93.9
Missing data	33	4.2
Total	791	100.0
Geographic Region (US)	Frequency	Percent
Central	90	11.4
East	24	3.0
Great Lakes	170	21.5
North	48	6.1
Midwest	71	9.0
Northwest	72	9.1
South	249	31.5
West	60	7.6
Missing data	7	.9
Total	791	100.0

Skin Integrity of Cohort
The following tables (Table 2, Table 3) show point prevalence of incontinence-associated dermatitis (IAD) and sacral pressure ulcers (sPUs).

RESULTS CONTINUED

- 23.3% (184/791) had some stage of IAD
 - 26.6% (49/184) had IAD present on admission (POA)
 - 73.4% (135/184) had facility-acquired IAD
- 9.6% (76/791) had some stage of sPU
 - 48.7% (37/76) had sPU POA
 - 51.3% (39/76) had facility-acquired sPU (fa-IAD)

Table 2. Point Prevalence of Incontinence and IAD

Point prevalence incontinence (N=791)		
Incontinence	Frequency	Percent
Present	426	53.9
Absent	333	42.1
Missing data	32	4.0
Type of incontinence (n=426)		
Urine	37	4.7
Fecal	135	17.1
Both	254	32.1
Missing data	32	4.0
Point prevalence IAD (n=426)		
IAD	Frequency	Percent
Present	184	43.2
Absent if incontinent	221	51.9
Missing data	21	4.9
Severity of IAD (n=184)		
Mild	120	65.2
Moderate	47	25.5
Severe	17	9.2
Missing data	242	30.6
Presence of fungal (n=426)		
Present	39	9.2
Absent	387	90.8
Present on admission (POA) or Facility Acquired (FA) (n=184)		
IAD-POA	49	26.6
FA-IAD	135	73.4
Total	184	100.0

RESULTS CONTINUED

Table 3. Point Prevalence of Sacral PUs

Point prevalence sacral PUs (N=791)		
Present	75	9.5
Absent	688	87
Missing data	28	3.5
Stage of sacral PUs (n=76)		
Stage	Frequency	Percent
Stage I	37	49.3
Stage II	21	28
Stage III	6	8
Stage IV	3	4
Unstageable	5	6.7
DTI	4	4
POA	37	48.7
Facility-Acquired	39	51.3

sPU Thickness/Incontinence Cross-Tabulation					
Count		URINE	FECAL	DOUBLE	Total
sPU Thickness	PARTIAL	2	22	34	58
	FULL	1	6	7	14
	DTI	0	3	1	4
Total		3	31	42	76

*DTI=deep tissue injury

Due to the large amount of data analyses performed on these data, all calculations were not presented in this poster. The summary statement is based on expert review of all analyses.

SUMMARY

Acute care patients with urinary, fecal or dual incontinence are vulnerable to IAD. All of the hospitals monitored in this survey used the traditional incontinence care method, which includes a multi-step approach. The prevalence of incontinence in this geographically diverse sample was 54% (426/791), and of these patients, 43% (184/426) had IAD. Approximately 73% of IAD cases were hospital acquired. IAD occurred in patients with urinary, fecal and dual incontinence; patients with fecal or dual incontinence were more likely to experience IAD, but the type of incontinence did not predict the severity of IAD. These findings are consistent with prior studies of IAD prevalence in the acute care setting that demonstrated an IAD prevalence varying from 20% to 27%[1,2]: however, these findings show that of incontinent patients 43% developed IAD. Study findings also demonstrate that urinary, fecal and dual incontinence are etiologic factors in the development of IAD.

Incontinence and IAD have also been linked to other forms of skin damage, such as sacral pressure ulcers. The relationship between IAD and development of sacral PU is not entirely understood; IAD is hypothesized to act as a risk factor for pressure ulcer development by impairing the skin's tolerance to pressure and shear[3]. While findings of this study confirm this association, further research is needed to more clearly elucidate the relationship between pressure formation and IAD in the acute care patient.

REFERENCES

1. Junkin J, Moore-Lisi I, Selekof JL. What we don't know can hurt us: pilot prevalence survey of incontinence and related perineal skin injury in acute care [Abstract]. WOCN 2005 National Conference Las Vegas NV.
2. Junkin J, Selekof JL. Prevalence of incontinence and associated skin injury in the acute care inpatient. Journal of Wound, Ostomy and Continence Nursing 2007; 34(3): 260-9.
3. Gray M., Beeckman D, Bliss DZ, Fader M, Logan S, Junkin J, Selekof J, Doughty D. Kurz P. Incontinence-Associated Dermatitis: A comprehensive review and update. Journal of Wound, Ostomy and Continence Nursing 2012; 39(1): 61-74.

21978